1625

The utility of AI-powered spatial classification of intratumoral CD8+ immune-cell topology in predicting overall survival in patients with melanoma as part of the CheckMate 067 clinical trial

George Lee,[1] Keyur Desai,[2] Hao Tang,[1] Daniel N. Cohen,[3] Scott Ely,[3] John B. Wojcik,[3] Jimena Trillo-Tinoco,[3] Benjamin J. Chen,[3] Benjamin Glass,[4] Nishant Agrawal,[4] Akshita Gupta,[1] Daniel J. Tenney,[3] Michael Montalto,[4] Vipul Baxi,[1] Robin Edwards,[3] Megan Wind-Rotolo[3]*

[1]Informatics and Predictive Sciences, Bristol Myers Squibb, Princeton, NJ, USA; [2]Global Biometric and Data Sciences, Bristol Myers Squibb, Princeton, NJ, USA; [3]Translational Medicine, Bristol Myers Squibb, Princeton, NJ, USA; [4]PathAI, Boston, MA, USA

*At the time this study was conducted

Scientific Content on Demand
To request a copy of this poster:

Scan QR code via a barcode reader application

QR codes are valid for 30 days after the congress presentation date.

Introduction

- The extent of cluster of differentiation 8-positive (CD8+) T-cell infiltration in the tumor microenvironment (TME) is a prognostic biomarker in patients with solid tumors[1-5]
- The spatial pattern and density of CD8+ T-cell infiltration (CD8 topology) can be categorized as "desert" (deficient in CD8+ cells), "excluded" (CD8+ cells at the tumor boundaries and surrounding stroma), and "inflamed" (CD8+ cells within the tumor parenchyma)[3,4,6,7]
- Understanding the association of CD8 topology with response to immune checkpoint inhibitors in the context of other key biomarkers, such as programmed death ligand 1 (PD-L1) expression, could help identify additional patients who may benefit from such therapies
- Classification of topology by standard manual pathology review is hampered by challenges with manual scoring of CD8 topology related to heterogeneous immune-cell infiltrates and interpathologist variability[5,6,8,9]
- Digital pathology approaches present an opportunity to quantify CD8 topology in a biologically meaningful, reproducible, and scalable way
- Using an artificial intelligence (AI)-driven algorithm, we retrospectively assessed the value of intratumoral CD8 topology together with PD-L1 expression as a composite biomarker of response to immunotherapy in patients with advanced melanoma in the phase 3 CheckMate 067 clinical trial (NCT01844505)

Methods

- We utilized a deep-learning platform (PathAI, Boston, MA) to quantify CD8+ T cells and recognize parenchymal and stromal compartments of the TME, using cellular and tissue annotations via the PathAI pathologist network (**Figure 1**)
 - Samples were classified as desert (deficient in CD8+ cells), excluded (CD8+ cells restricted to stroma or invasive margin), and inflamed (CD8+ cells infiltrating tumor parenchyma)
 - The accuracy of the AI-predicted CD8+ cell counts was evaluated using the Frames Validation method, where model counts in 28 held-out test frames from 22 baseline whole slide images (WSIs) (obtained from patients with melanoma enrolled in the CheckMate 067 trial) were compared with a consensus manual count from 5 independent pathologists using Pearson correlation

Figure 1. Overview of development of the AI algorithm to predict CD8 topology

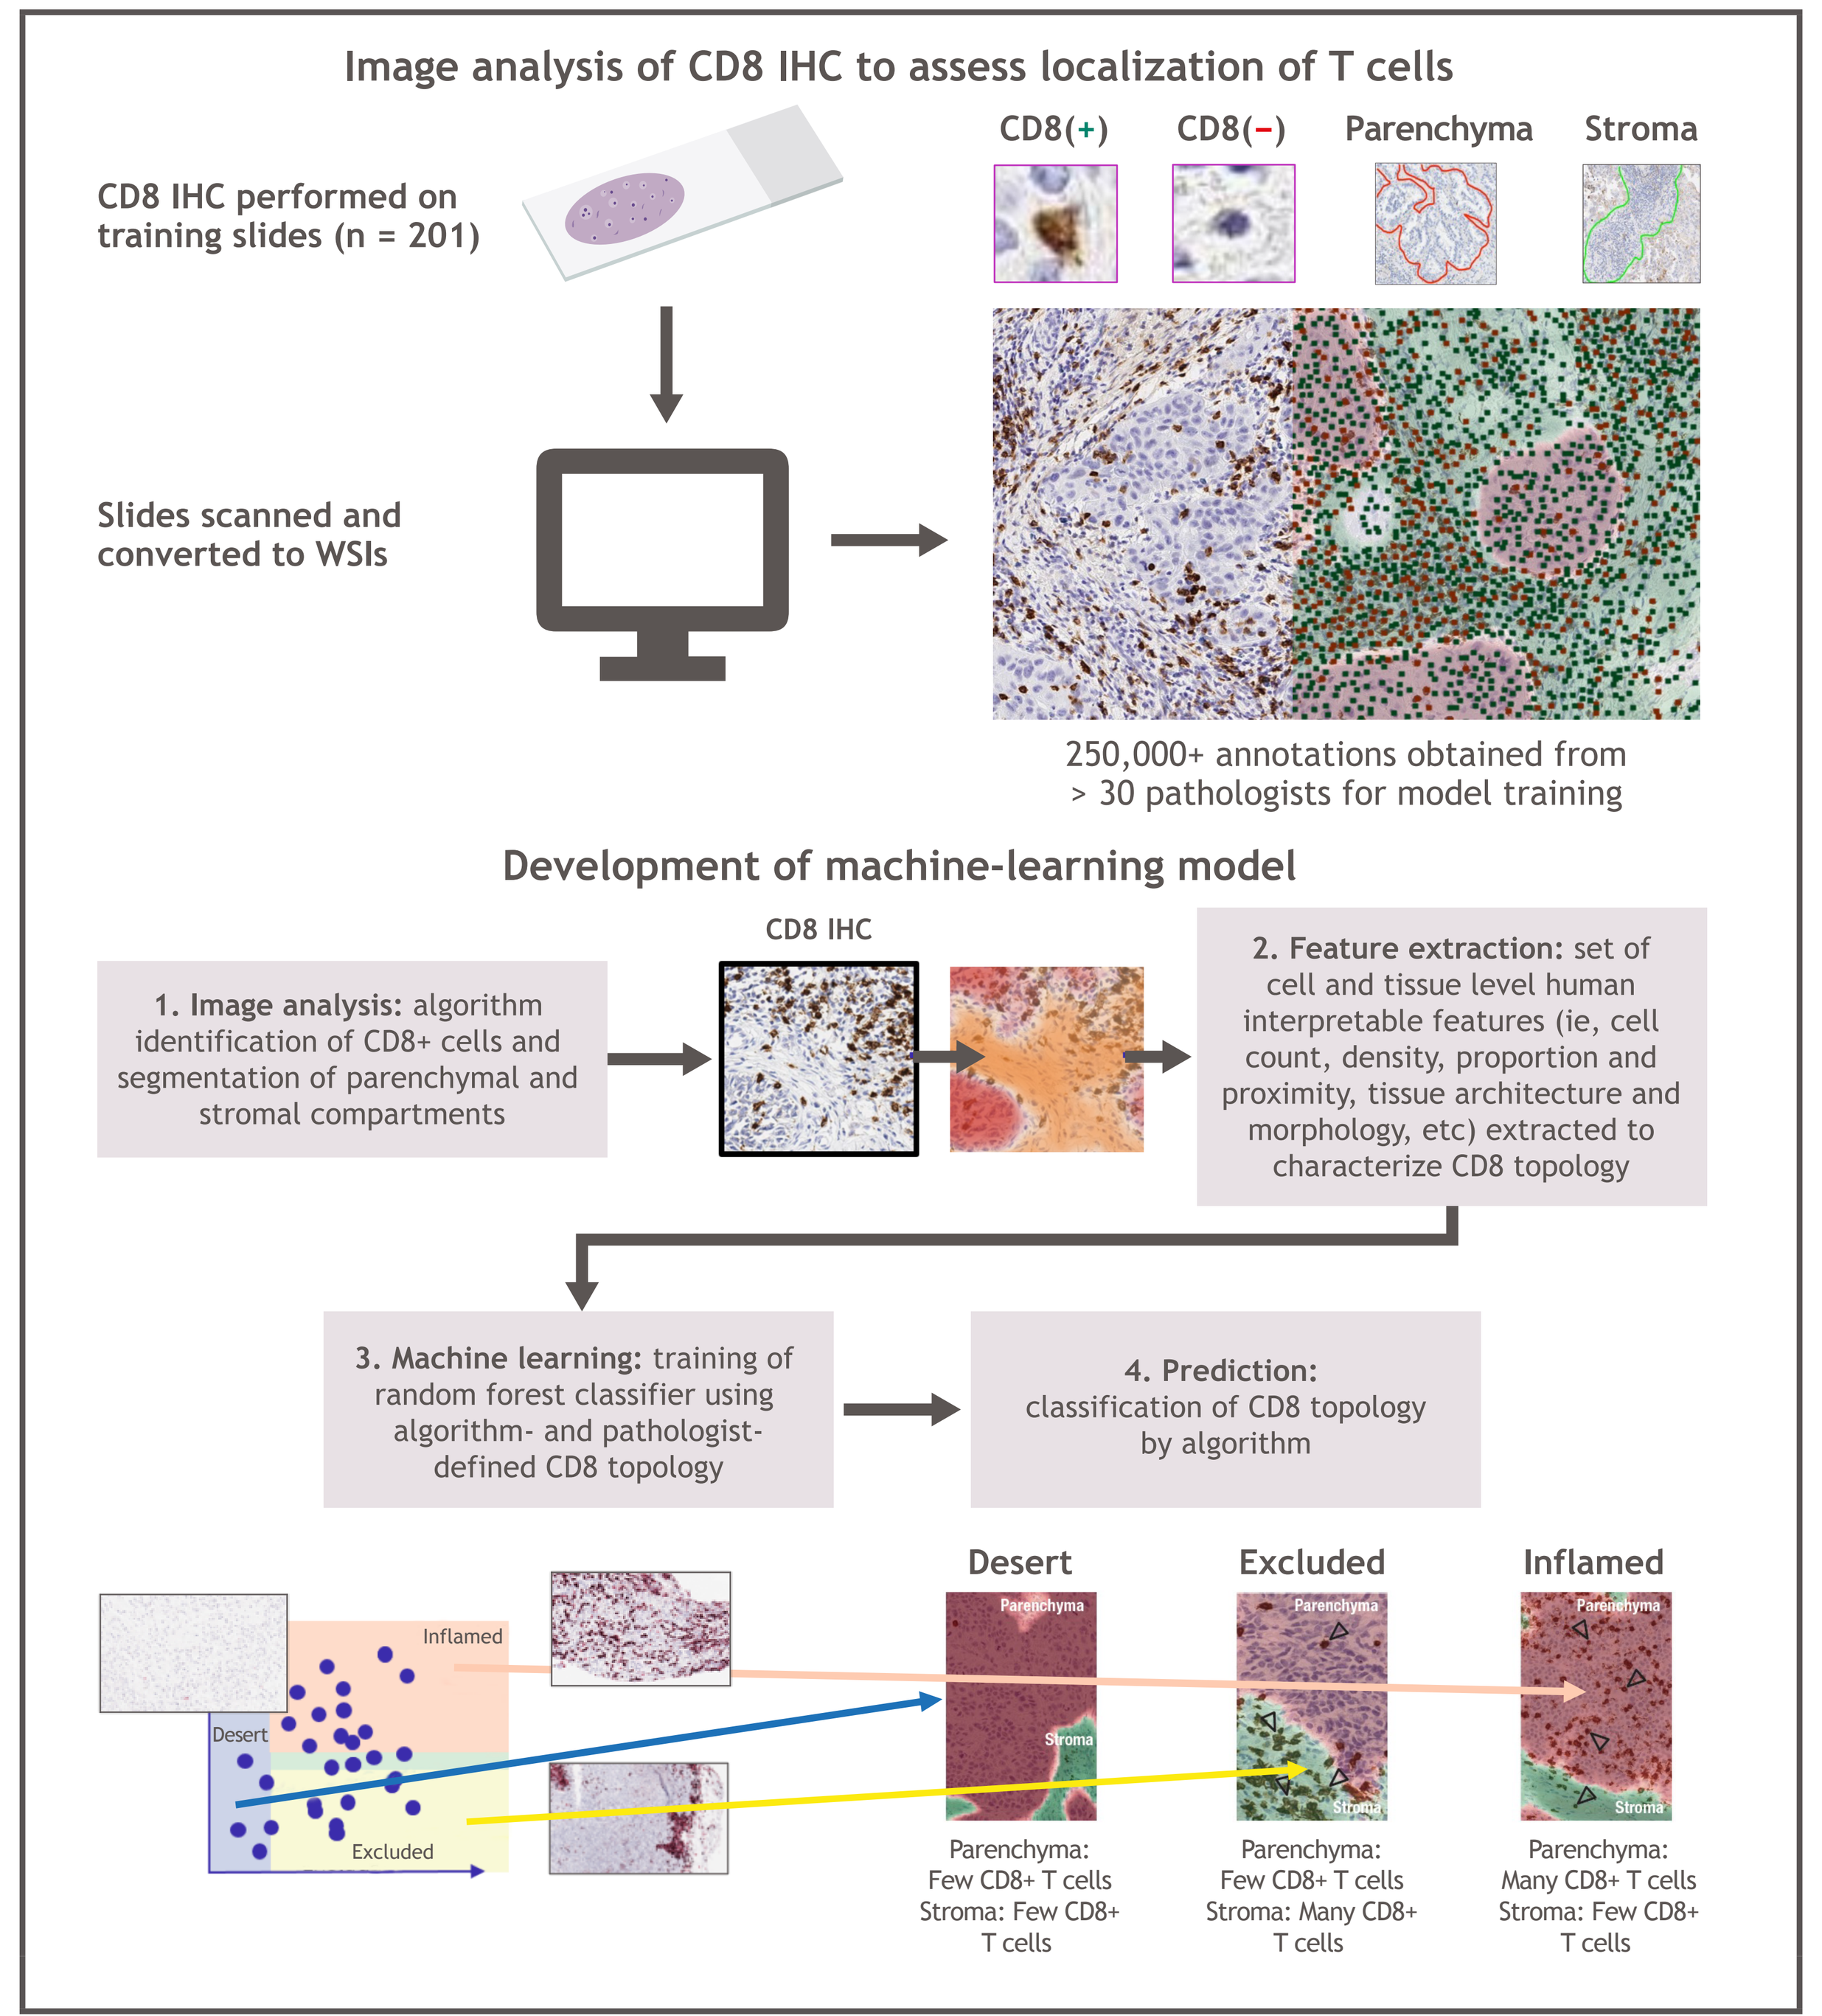
Image analysis of CD8 IHC to assess localization of T cells
CD8 IHC performed on training slides (n = 201)
CD8(+)
CD8(−)
Parenchyma
Stroma
Slides scanned and converted to WSIs
250,000+ annotations obtained from > 30 pathologists for model training
Development of machine-learning model
CD8 IHC
1. Image analysis: algorithm identification of CD8+ cells and segmentation of parenchymal and stromal compartments
2. Feature extraction: set of cell and tissue level human interpretable features (ie, cell count, density, proportion and proximity, tissue architecture and morphology, etc) extracted to characterize CD8 topology
3. Machine learning: training of random forest classifier using algorithm- and pathologist-defined CD8 topology
4. Prediction: classification of CD8 topology by algorithm
Inflamed
Desert
Excluded
Desert
Excluded
Inflamed
Parenchyma: Few CD8+ T cells Stroma: Few CD8+ T cells
Parenchyma: Few CD8+ T cells Stroma: Many CD8+ T cells
Parenchyma: Many CD8+ T cells Stroma: Few CD8+ T cells

- The features extracted from these annotations were used to train a random forest classifier to predict CD8 topology using parenchymal and stromal CD8+ immune-cell measurements
- To validate AI-predicted CD8 topology classifications, pathologists manually classified CD8 topology on an independent set of digital WSIs from 40 CD8 immunohistochemistry (IHC) slides (C8/144B, Agilent, Santa Clara, CA) obtained from resection/core needle biopsies of patients with advanced melanoma enrolled in the CheckMate 038 clinical trial
- Inter-pathologist concordance was compared with concordance between pathologists and the AI algorithm in classifying CD8 topology
- We then generated a composite biomarker that consisted of AI-classified CD8 topology combined with PD-L1 expression on tumor cells (TCs) using a cutoff of 1% as a reference (determined as part of the original CheckMate 067 trial[10])
- Associations between overall survival (OS) in patients with previously untreated metastatic melanoma who received nivolumab plus ipilimumab (NIVO + IPI; n = 102), NIVO monotherapy (n = 107), or IPI monotherapy (n = 106) in CheckMate 067, and AI-defined CD8 topology alone or in combination with PD-L1 expression were estimated using Cox proportional hazards models
- PD-L1 expression on TCs was determined using the Dako PD-L1 IHC 28-8 pharmDx assay (Agilent, Santa Clara, CA)
 - Manual scoring of PD-L1 expression on TCs was derived from the percentage of TCs with complete or partial PD-L1 expression at any level of intensity divided by the number of total TCs
 - Tumor samples with PD-L1 expression ≥ 1% were classified as PD-L1–positive, and those with PD-L1 expression < 1% were classified as PD-L1–negative
- Kaplan-Meier curves were used to illustrate comparisons of survival in samples identified as immune-excluded or -inflamed and/or PD-L1-positive or PD-L1-negative
- Patients with an CD8-desert phenotype were not included in the survival analyses due to low sample size

Results

- The correlation between AI-powered and pathologist quantification of CD8+ cells was high (**Figure 2**)
- Classifications of CD8 topology by the AI algorithm were concordant with manual scoring performed and determined by a consensus of pathologists (Cohen's κ, 0.79) and non-inferior to the agreement between 2 pathologists (Cohen's κ, 0.65)

Figure 2. AI categorization of CD8+ cell counts compared with a consensus of pathologists

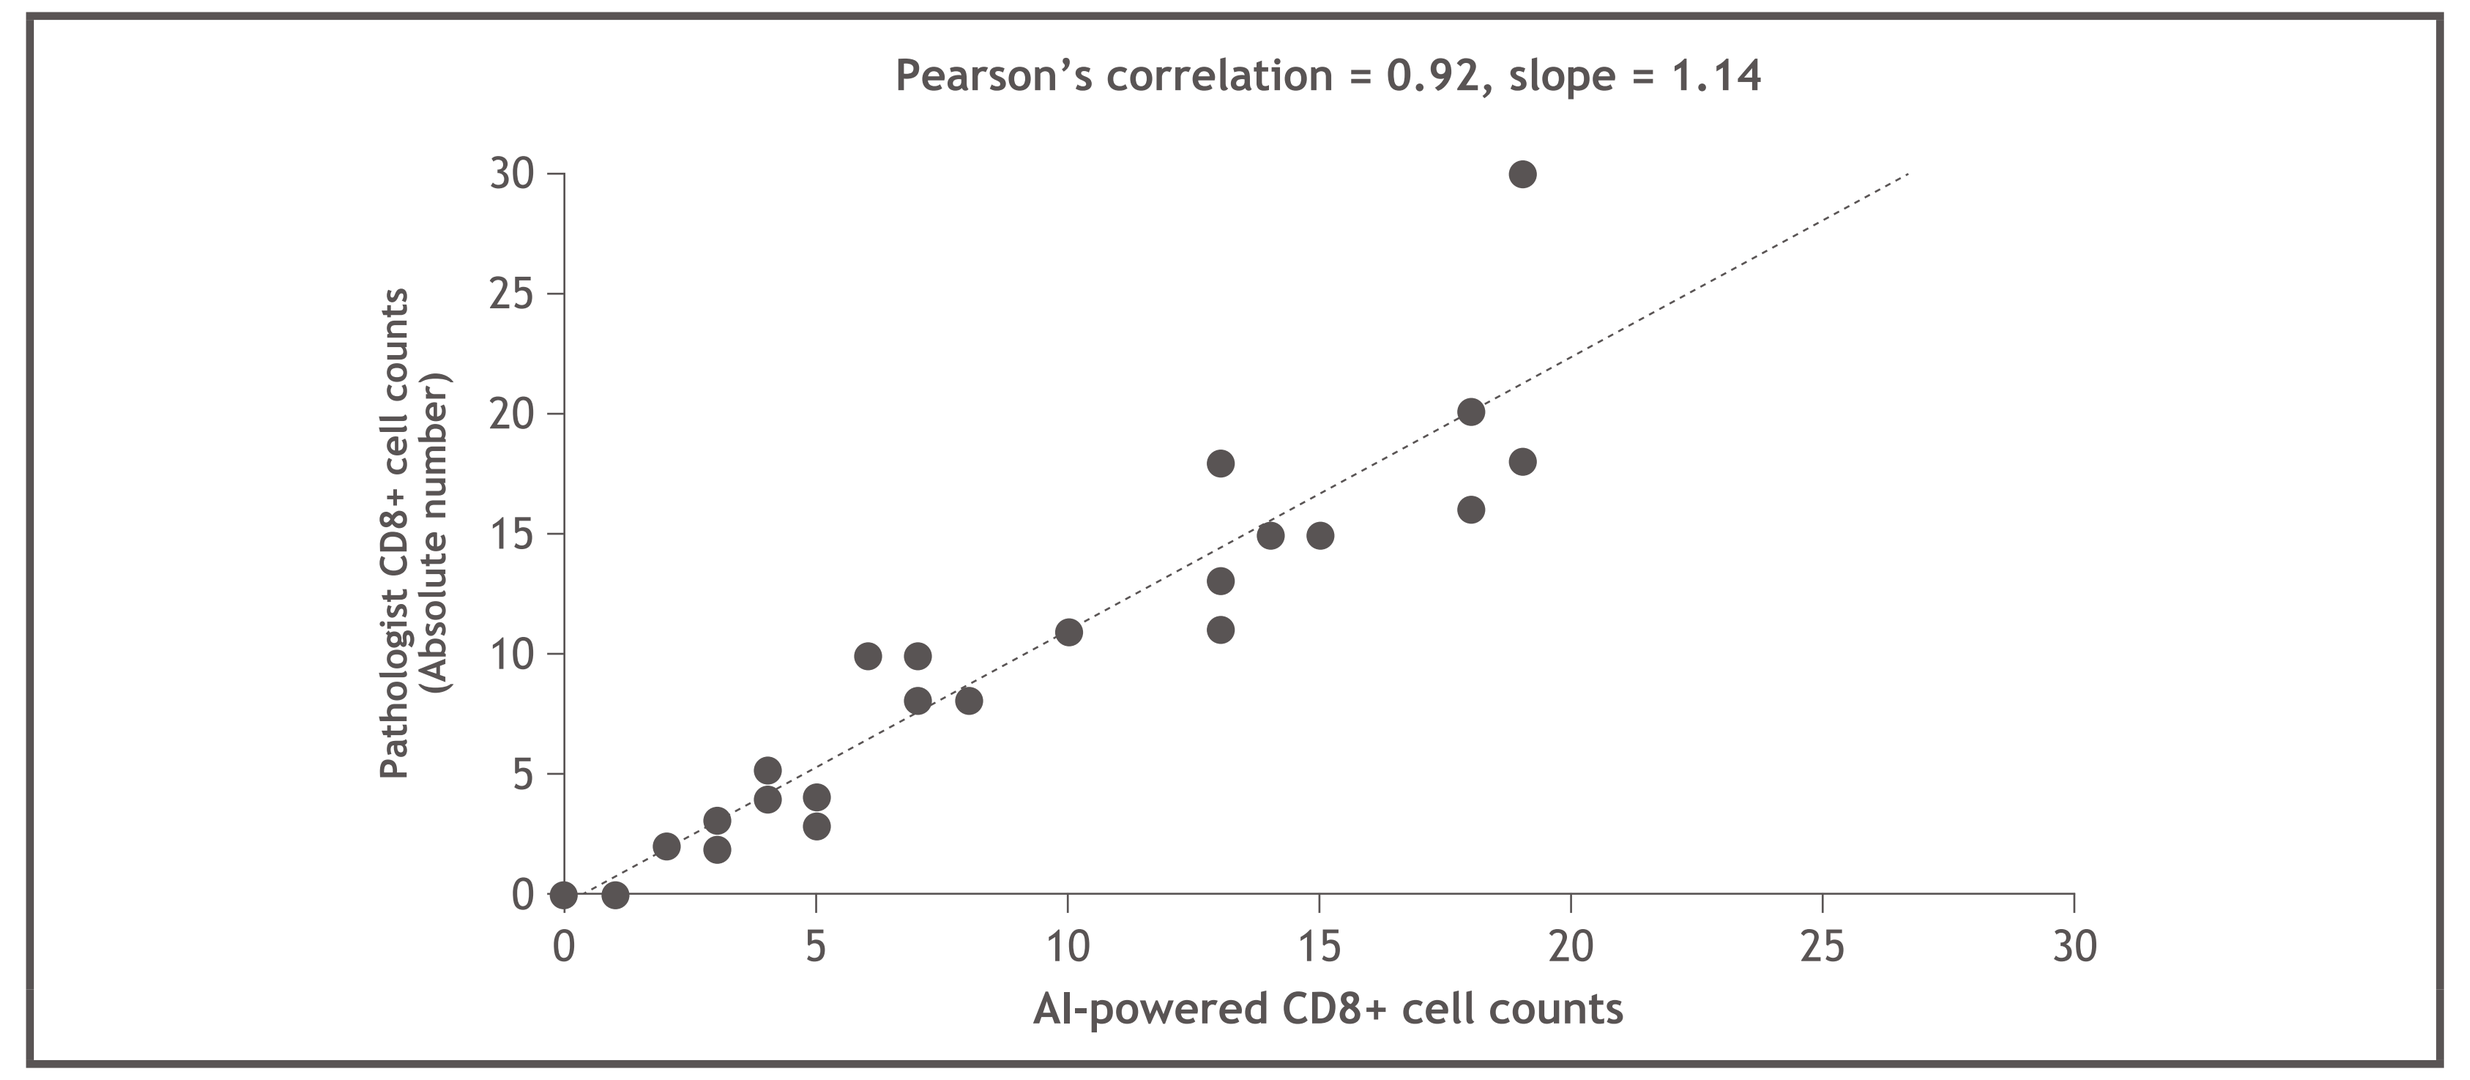
Pearson's correlation = 0.92, slope = 1.14
Pathologist CD8+ cell counts (Absolute number)
AI-powered CD8+ cell counts

- Measurements of the percentage of CD8 positivity in the stromal and parenchymal compartments were similar to AI-predicted CD8 categorizations (**Figure 3**)
 - Pathologist review of AI-predicted topology found these categorizations to be accurate

Figure 3. AI categorization of CD8 topology compared with intratumoral CD8 measurements

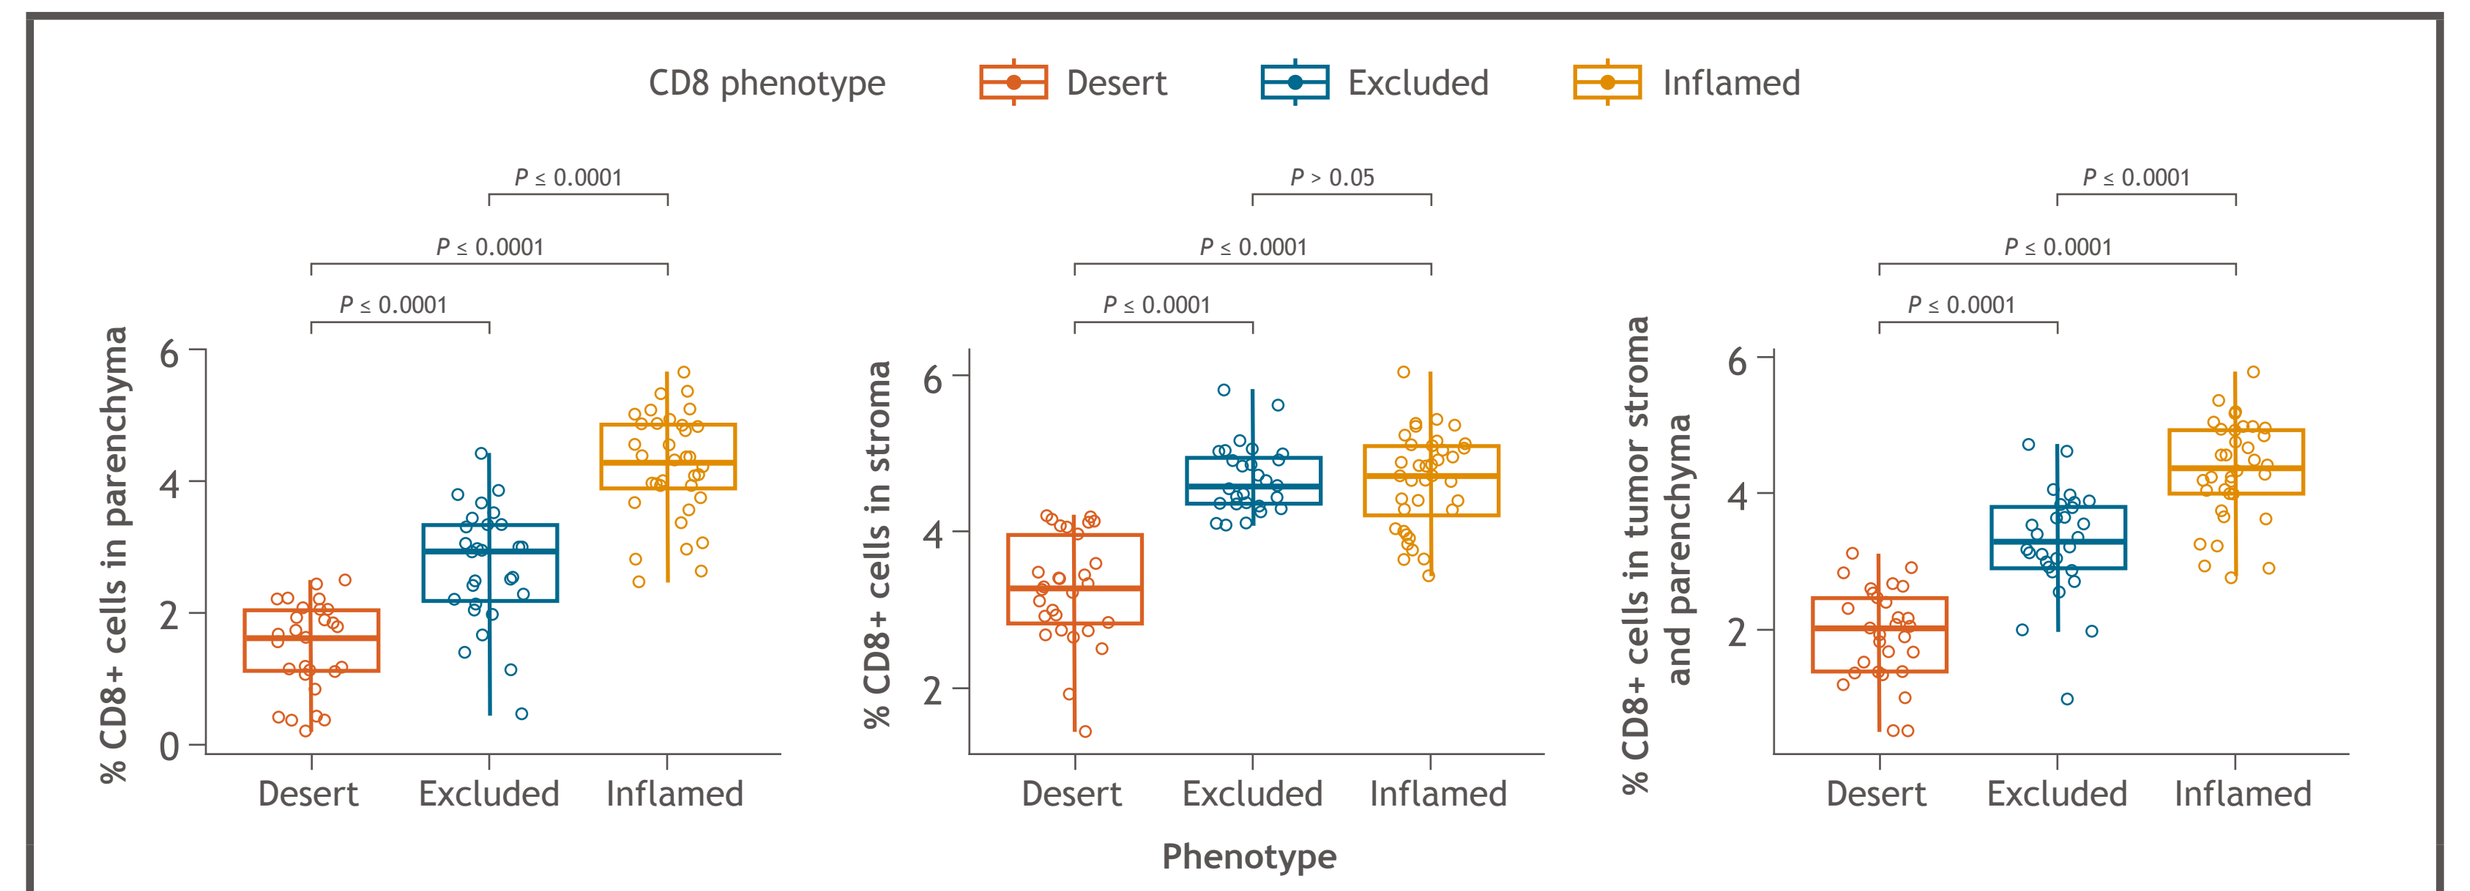
CD8 phenotype
Desert
Excluded
Inflamed
% CD8+ cells in parenchyma
% CD8+ cells in stroma
% CD8+ cells in tumor stroma and parenchyma
Phenotype

Desert: deficient in CD8+ cells. Excluded: CD8+ cells restricted to stroma or invasive margin. Inflamed: CD8+ cells infiltrating tumor parenchyma.

- Of 105 patients with PD-L1 expression ≥ 1%, 36 (34%) had phenotypes that were classified as CD8-excluded and 60 (57%) had phenotypes that were classified as CD8-inflamed (**Figure 4**)
- Of 104 patients with PD-L1 expression < 1%, 40 (38%) had phenotypes that were classified as CD8-excluded and 27 (26%) had phenotypes that were classified as CD8-inflamed
 - Due to the low numbers of patients with PD-L1 expression ≥ 1% who had phenotypes classified as CD8-desert (9%), it was not possible to perform the OS analysis with this group

Figure 4. Classification of patients by CD8 topology and TC PD-L1 expression

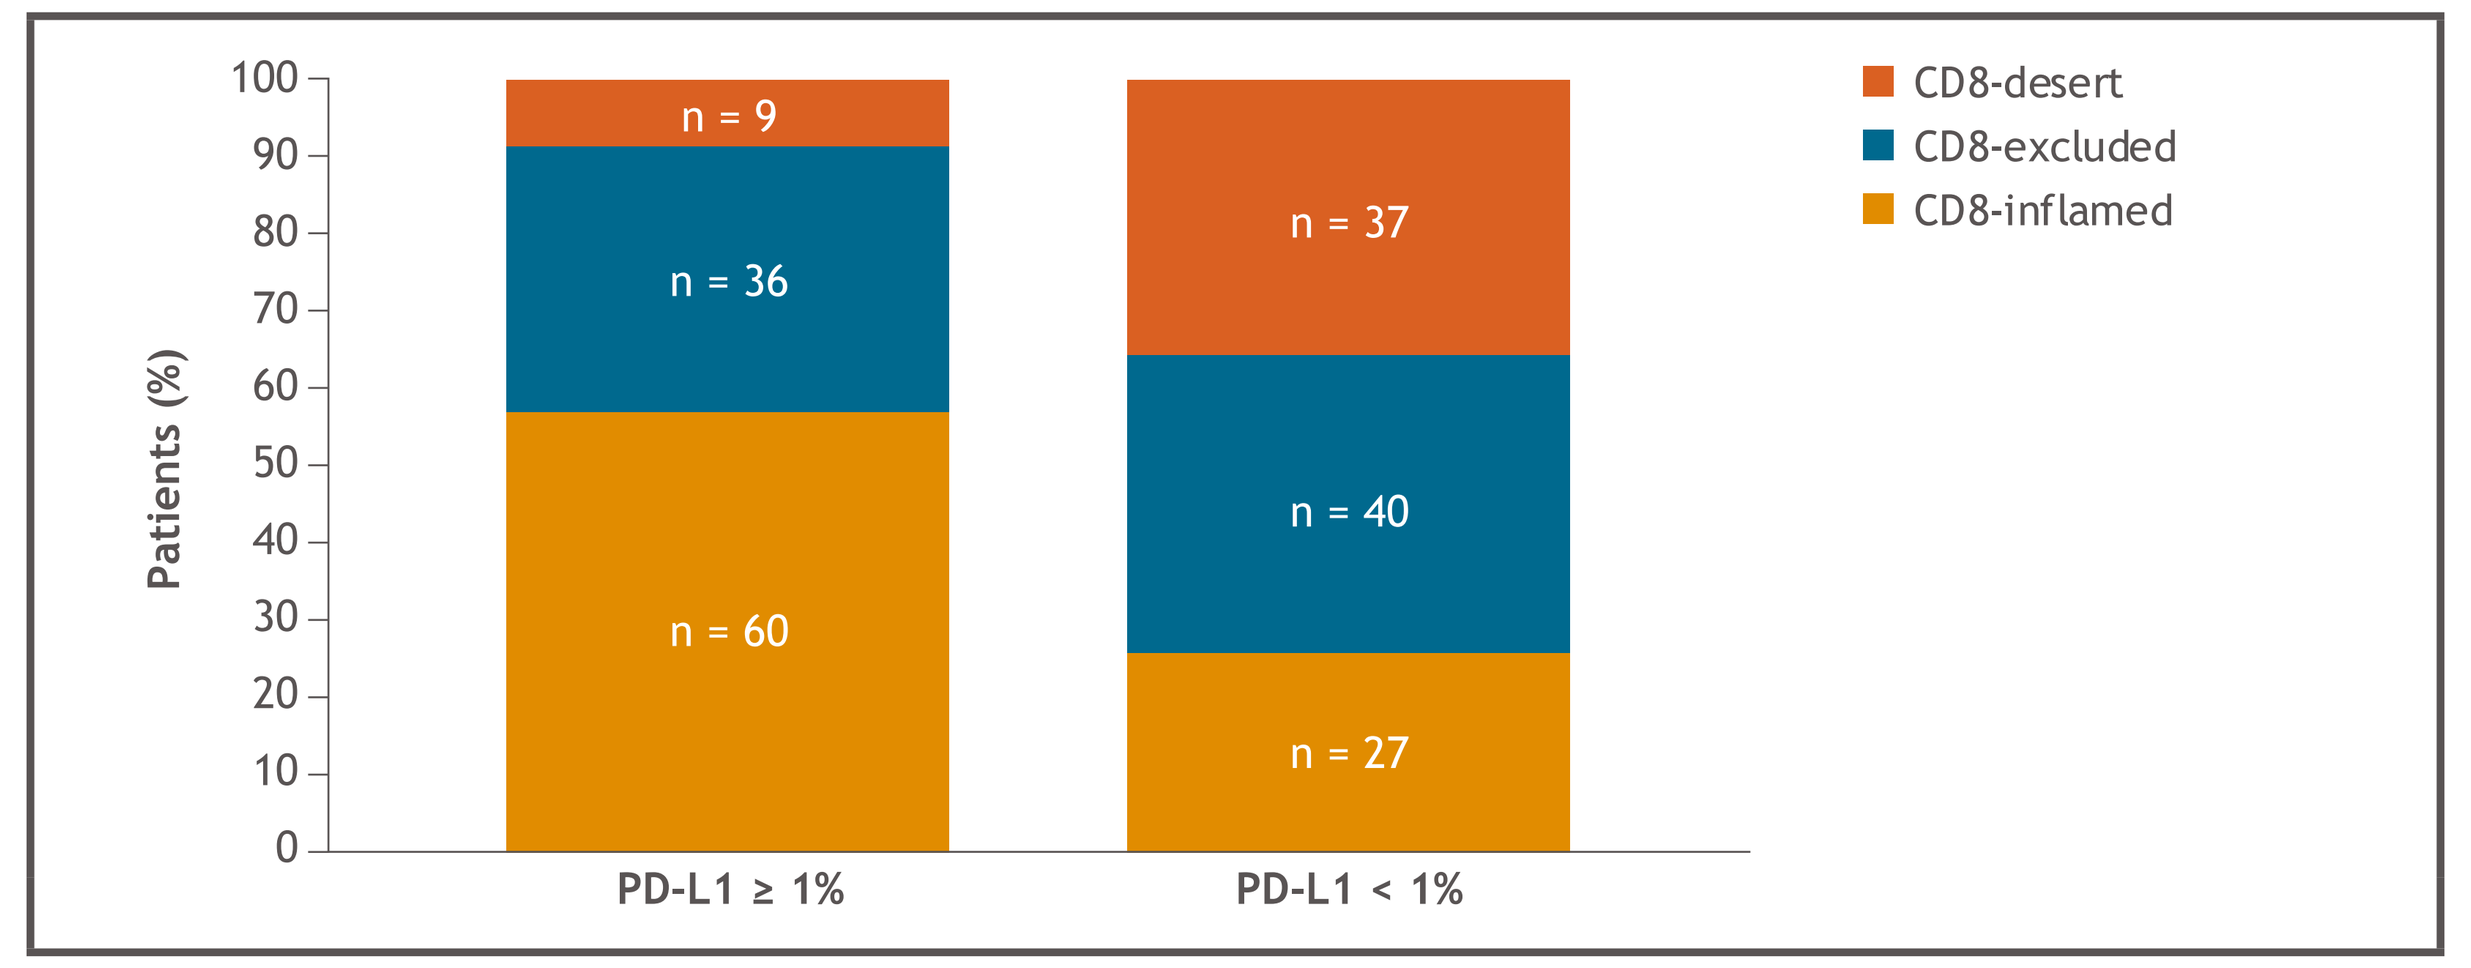
CD8-desert
CD8-excluded
CD8-inflamed
Patients (%)
n = 9
n = 36
n = 60
n = 37
n = 40
n = 27
PD-L1 ≥ 1%
PD-L1 < 1%

- In patients with PD-L1 expression ≥ 1%, no statistically significant difference in OS was observed between the CD8-excluded and CD8-inflamed phenotypes across the different treatment arms (**Figure 5**)
- Among patients with PD-L1 expression < 1%, those with a CD8-excluded phenotype benefited significantly from NIVO + IPI therapy (P = 0.002) compared with those with a CD8-inflamed phenotype, whereas the difference between phenotypes was not significant for those treated with NIVO monotherapy (P = 0.41) (**Figure 5, Table 1**)

Figure 5. OS by CD8 topology (inflamed vs excluded)[a] and PD-L1 expression (<1% vs ≥ 1%)

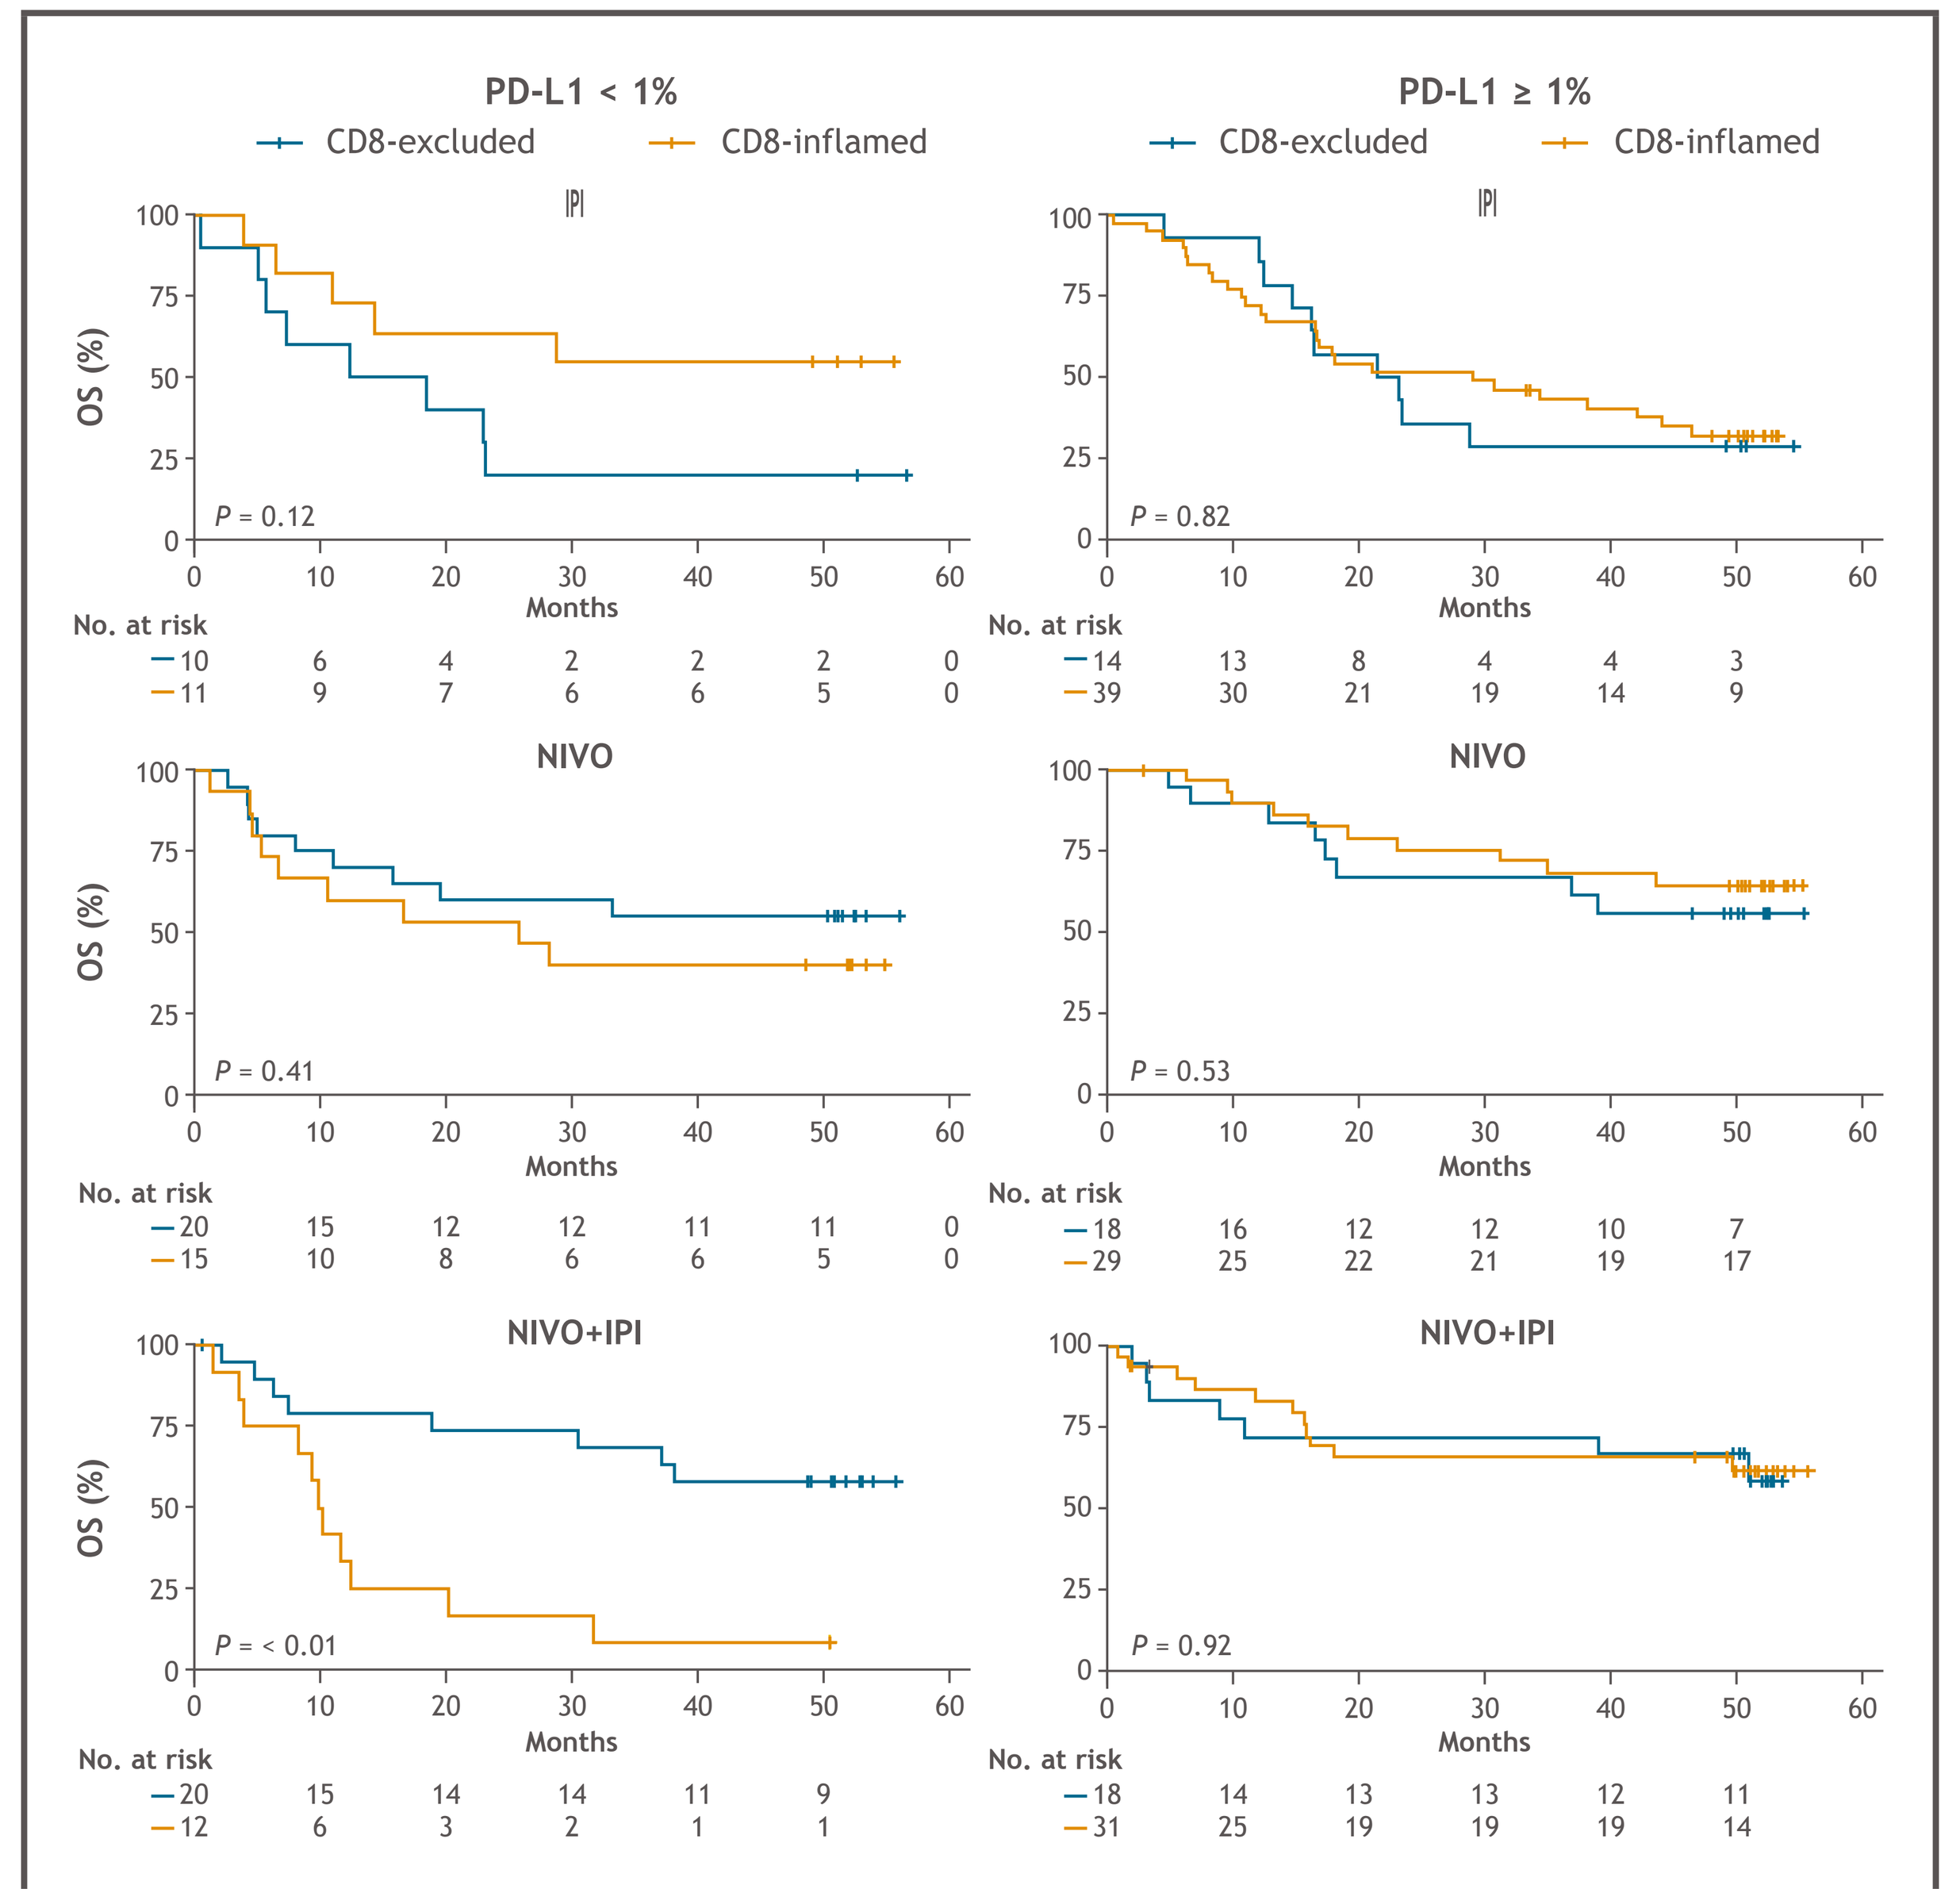
PD-L1 < 1%
PD-L1 ≥ 1%
CD8-excluded
CD8-inflamed
IPI
NIVO
NIVO+IPI
OS (%)
Months
No. at risk
P = 0.12
P = 0.82
P = 0.41
P = 0.53
P = < 0.01
P = 0.92

[a]Samples identified as CD8-desert not included due to small sample size.

Table 1. OS by CD8 topology in patients with PD-L1 expression < 1%

Treatment arm	IPI		NIVO + IPI		NIVO	
Phenotype (n)	PD-L1 < 1%, CD8-excluded (10)	PD-L1 < 1%, CD8-inflamed (11)	PD-L1 < 1%, CD8-excluded (20)	PD-L1 < 1%, CD8-inflamed (12)	PD-L1 < 1%, CD8-excluded (20)	PD-L1 < 1%, CD8-inflamed (15)
Median OS, months	15.4	> 50.0	> 50.0	10.1	> 50.0	25.8
HR (95% CI)	2.44 (0.79–7.56); P = 0.12		0.23 (0.09–0.61); P < 0.01		0.68 (0.27–1.70); P = 0.41	

HR, hazard ratio.

- In patients treated with NIVO + IPI with a CD8-inflamed phenotype and PD-L1 expression < 1%, a greater incidence of adverse events (AEs) occurred than in patients with an excluded phenotype (**Figure 6**); however, this is limited by a small sample size
- Among patients with PD-L1 expression < 1%, severe AEs (SAEs; defined as grade ≥ 3) were less frequent in patients with a CD8-excluded phenotype than in those with a CD8-inflamed phenotype (data not shown)

Figure 6. Association between tumor phenotype and AE incidence

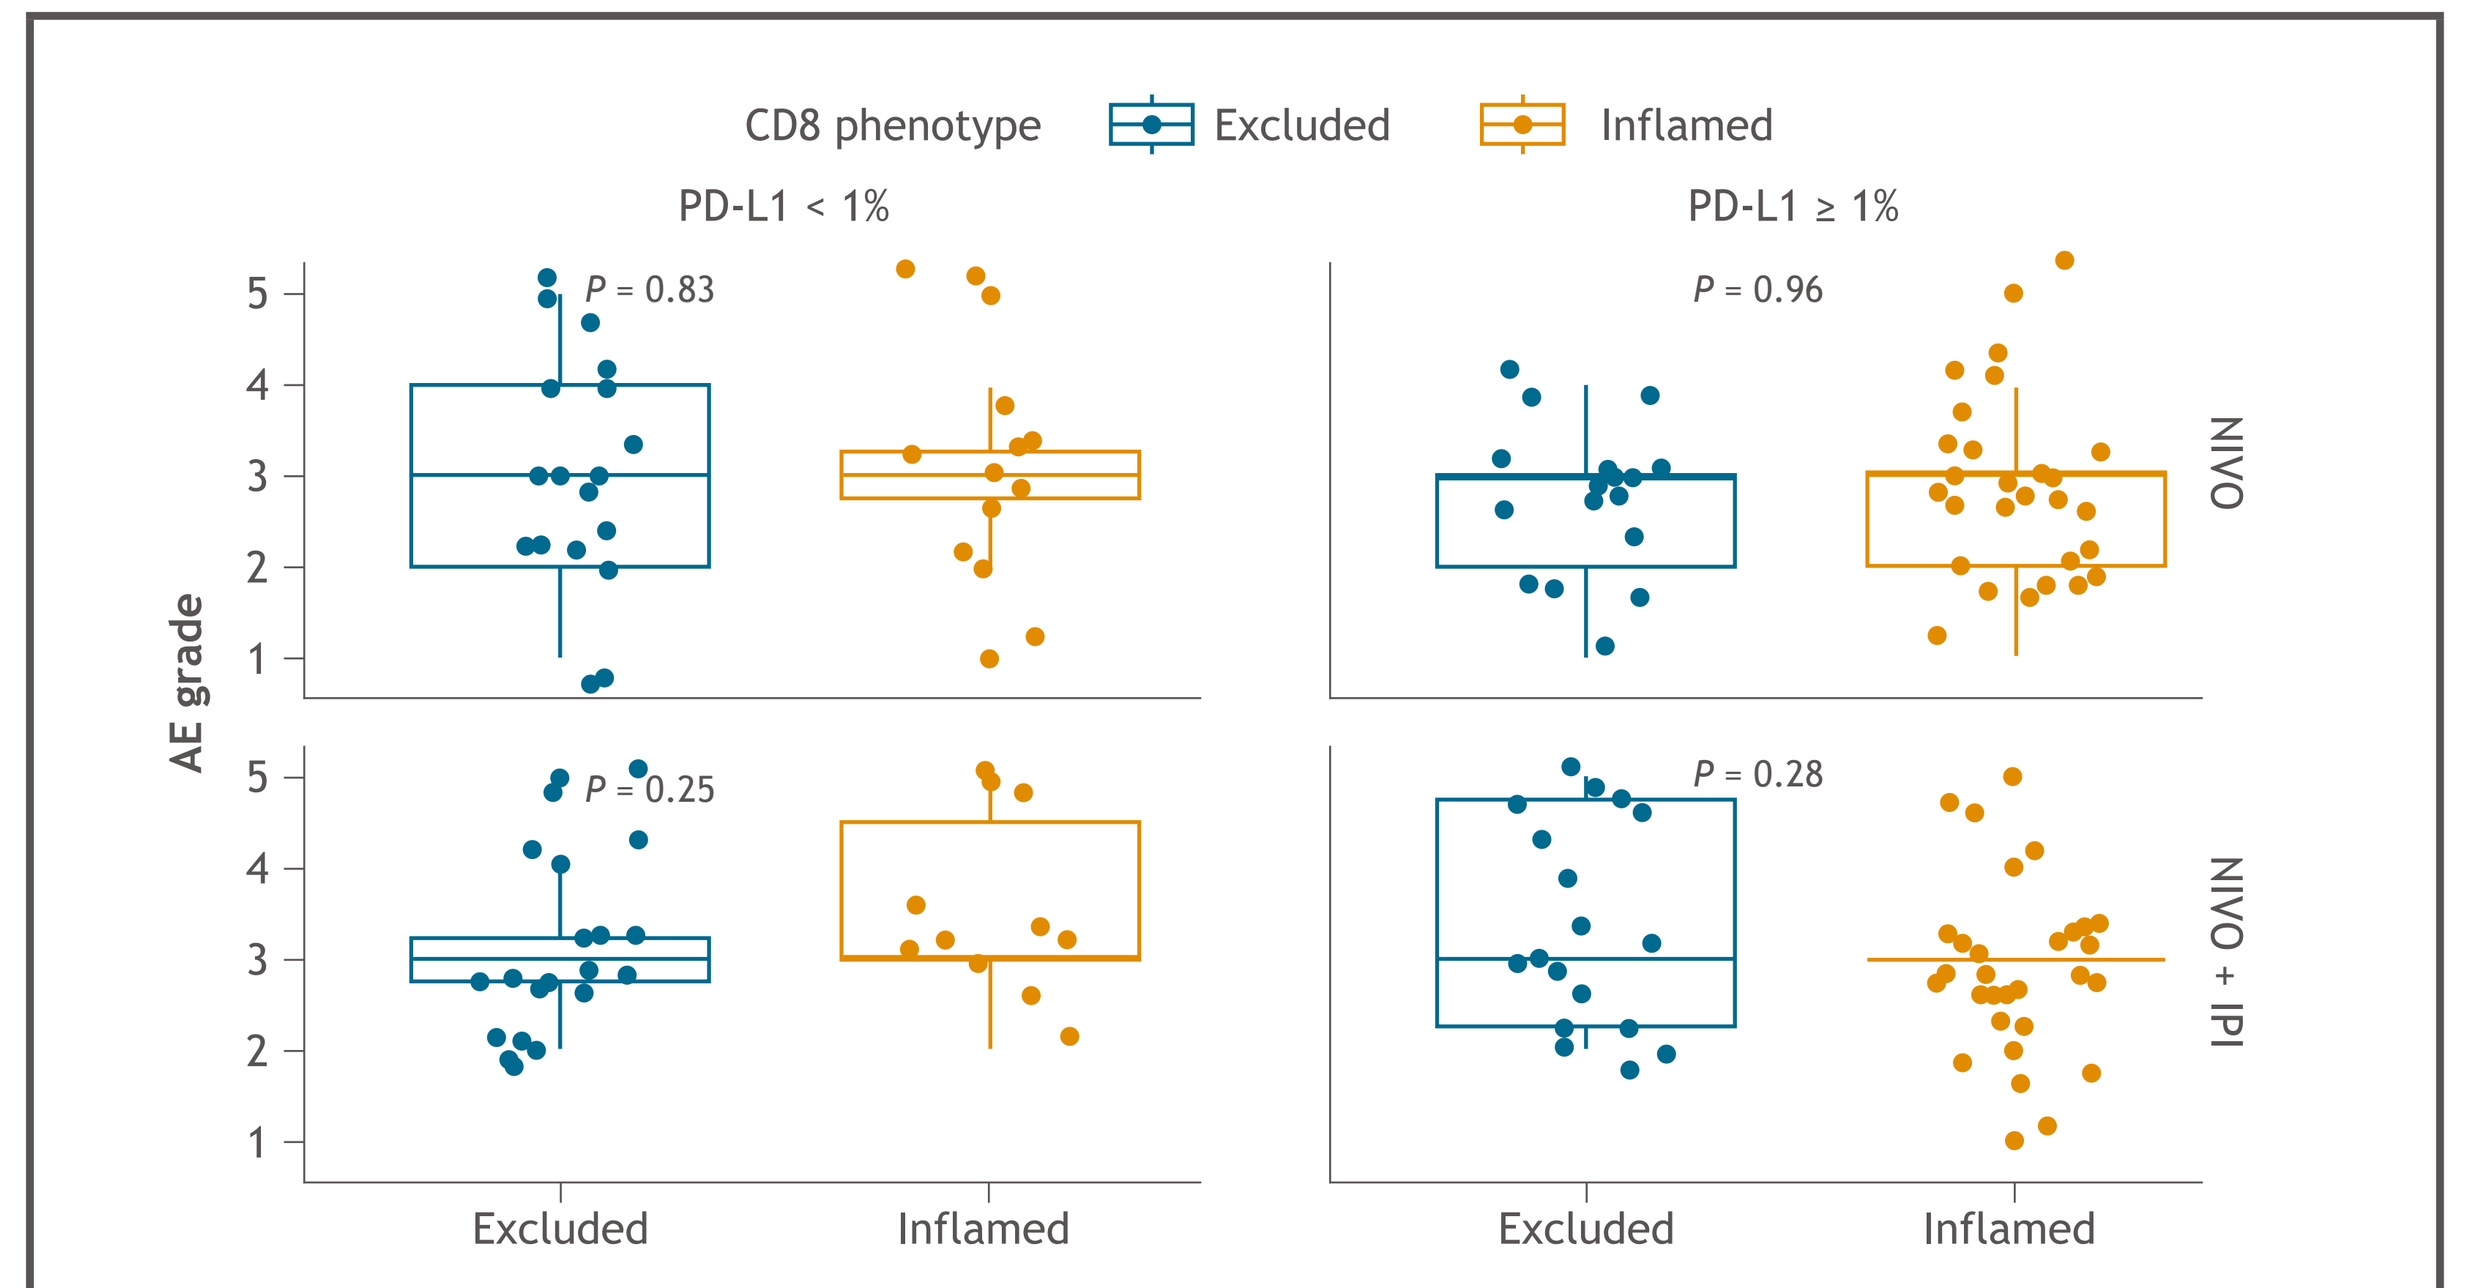
CD8 phenotype
Excluded
Inflamed
PD-L1 < 1%
PD-L1 ≥ 1%
P = 0.83
P = 0.96
P = 0.25
P = 0.28
AE grade
NIVO
NIVO + IPI
Excluded
Inflamed

Whiskers represent values 1.5× the upper and lower limits of the interquartile range.

- To investigate whether combining two sets of patients could result in greater clinical benefit for patients treated with NIVO ± IPI, we introduced a composite biomarker that combines patients with PD-L1 expression < 1% and classified as CD8-excluded phenotype and those with PD-L1 expression ≥ 1%, accounting for 68%–71% of tumors (**Table 2**)
 - Patients identified as positive based on the composite biomarker demonstrated a stronger association with survival than those identified as biomarker-negative (PD-L1 < 1%, and CD8-inflamed or CD8-desert) compared with using PD-L1 expression alone in both the NIVO + IPI and NIVO monotherapy groups (**Figures 7A and 7B**)

Table 2. HRs comparing OS in patients identified as biomarker-positive using an AI-driven composite biomarker compared with PD-L1 expression alone

Biomarker	PD-L1 ≥ 1%: Number of patients identified as positive (%)	PD-L1 ≥ 1%: OS HR[a] (95% CI)	Composite biomarker (PD-L1 ≥ 1%, or CD8-excluded and PD-L1 < 1%): Number of patients identified as positive (%)	Composite biomarker: OS HR[b] (95% CI)
NIVO + IPI (n = 102)	52 (51%)	0.50 (0.29–0.89); P = 0.017	72 (71%)	0.35 (0.20–0.61); P < 0.001
NIVO (n = 107)	53 (50%)	0.46 (0.27–0.79); P = 0.005	73 (68%)	0.37 (0.22–0.62); P < 0.001

[a]HRs represent patients with PD-L1 expression ≥ 1% compared with patients with PD-L1 < 1%; [b]HRs represent patients with PD-L1 ≥ 1%, or CD8-excluded and PD-L1 < 1% compared with patients with PD-L1 expression < 1% and CD8-inflamed or CD8-desert.

Figure 7. OS by PD-L1 expression or using composite biomarker in patients treated with NIVO + IPI (A) or NIVO monotherapy (B)

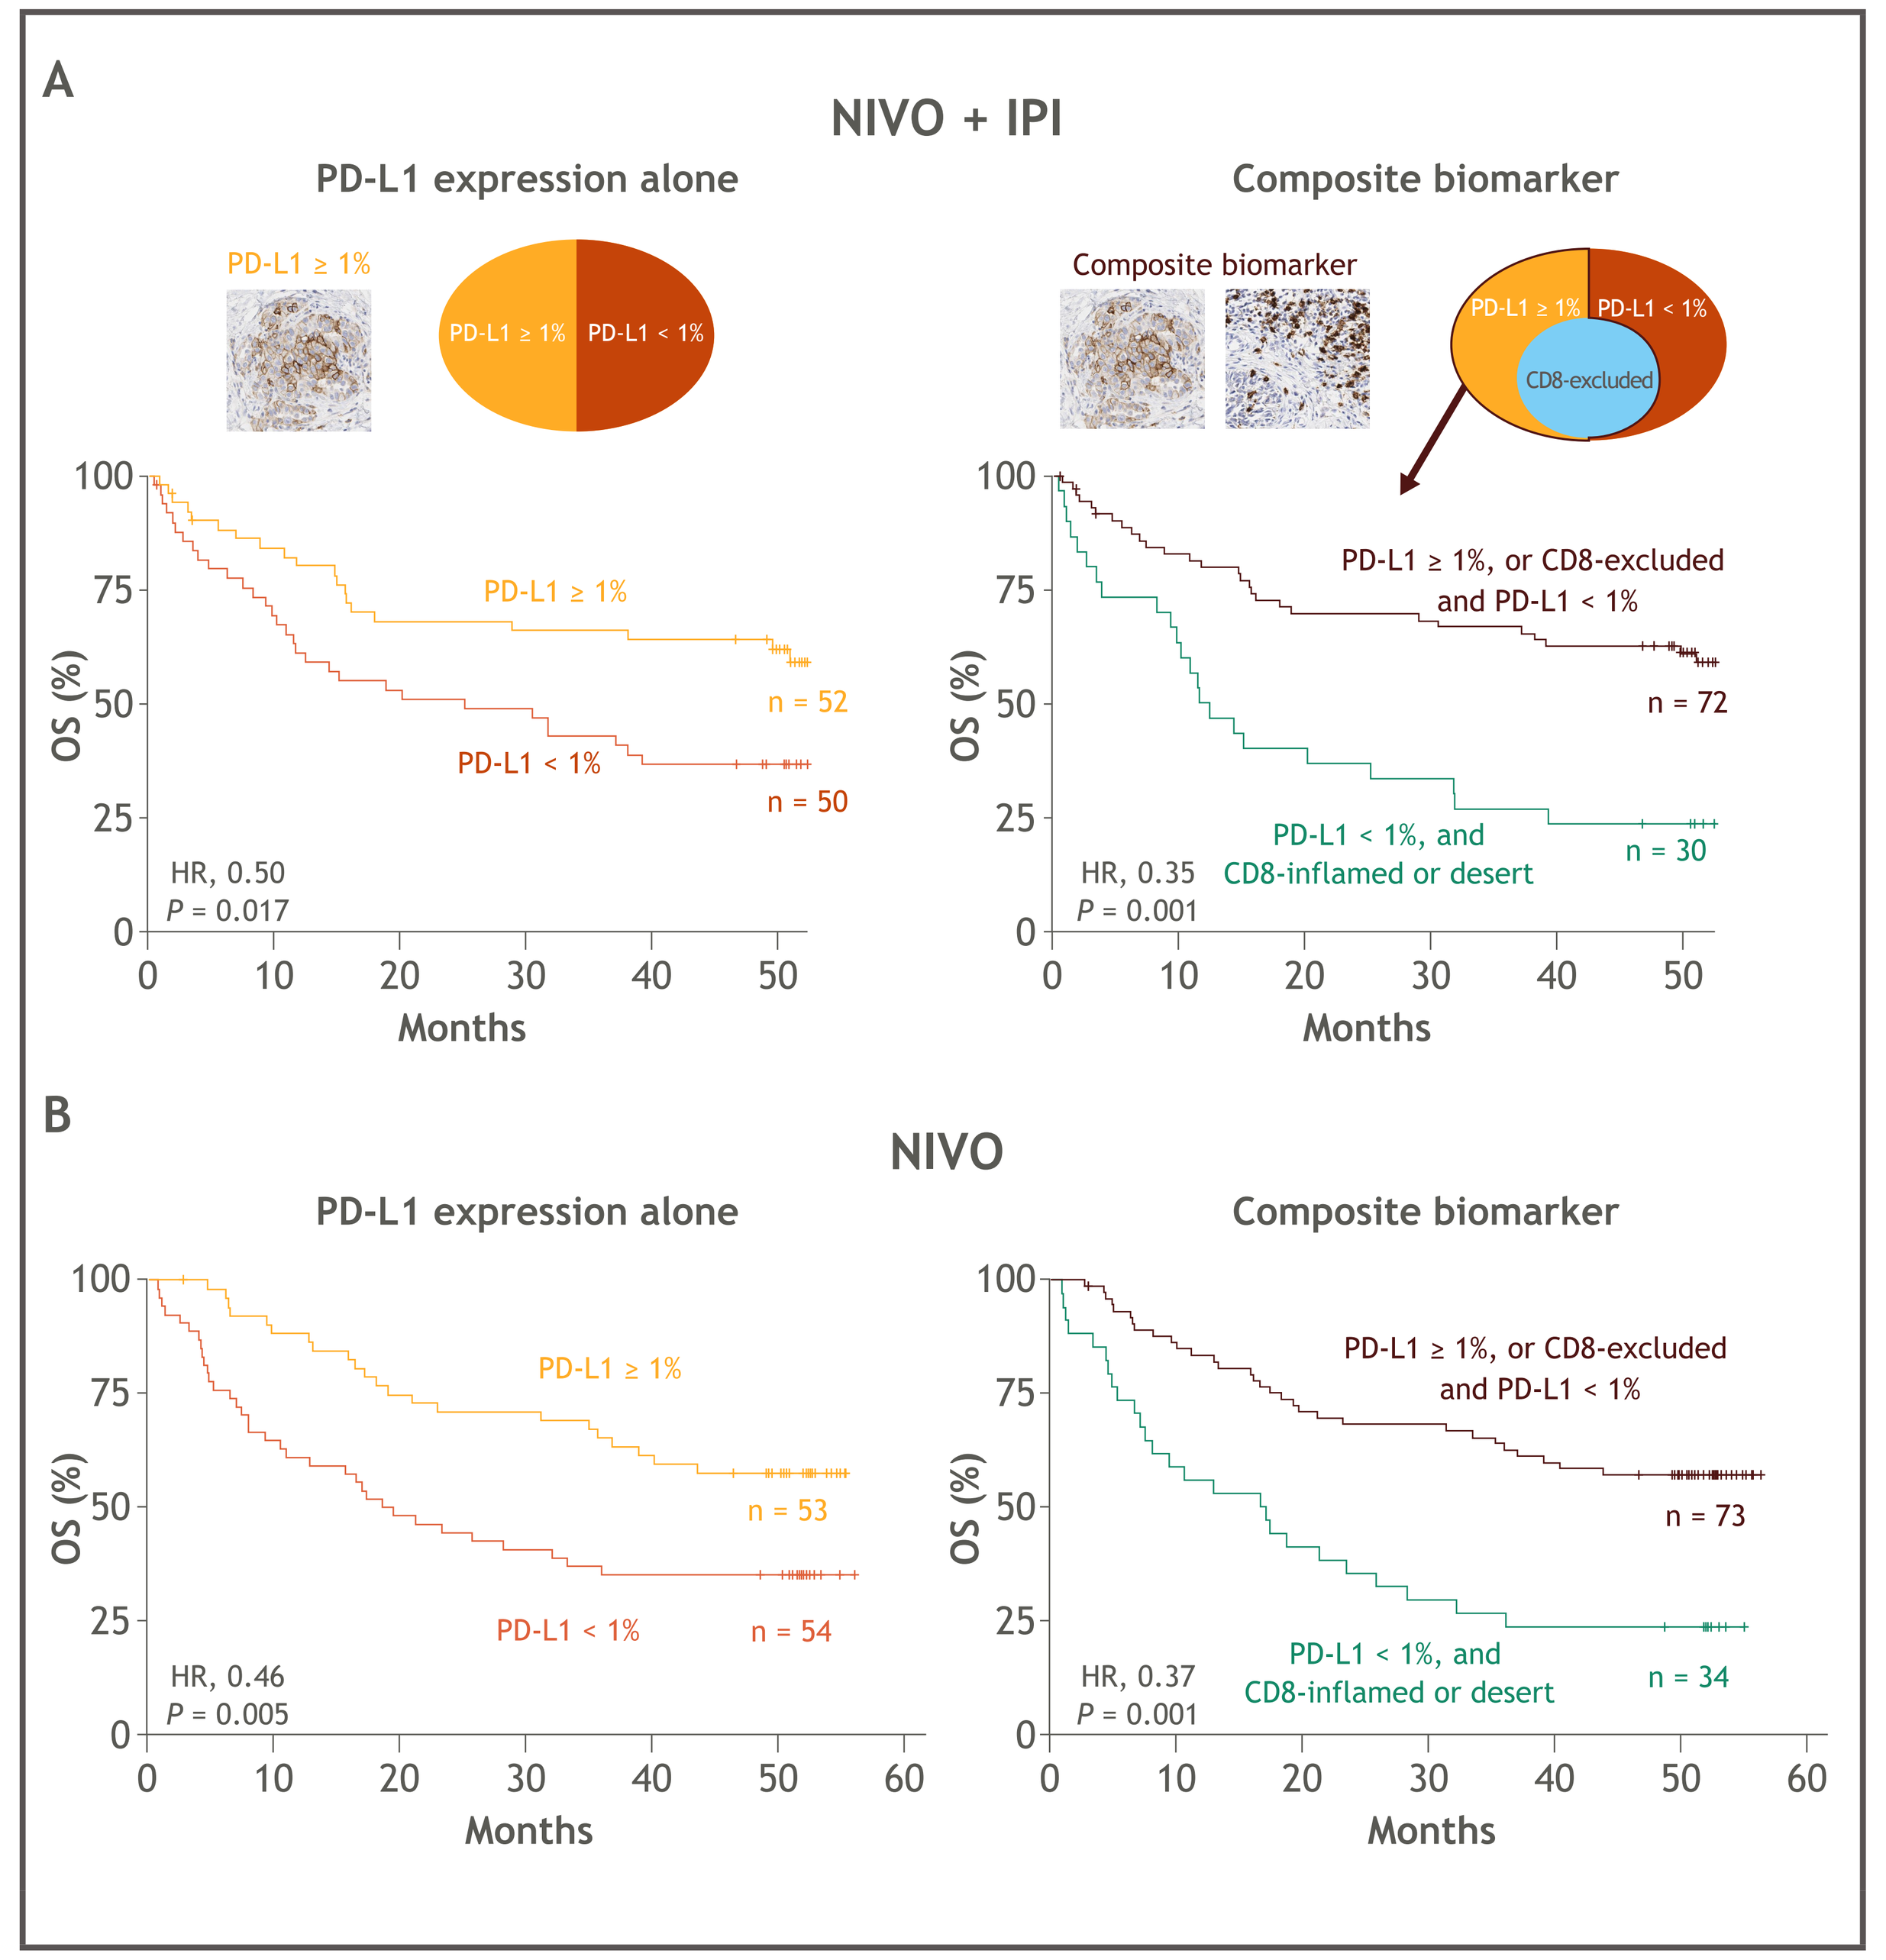
A
NIVO + IPI
PD-L1 expression alone
Composite biomarker
PD-L1 ≥ 1%
PD-L1 < 1%
CD8-excluded
PD-L1 ≥ 1%, or CD8-excluded and PD-L1 < 1%
PD-L1 < 1%, and CD8-inflamed or desert
n = 52
n = 50
n = 72
n = 30
HR, 0.50
P = 0.017
HR, 0.35
P = 0.001
B
NIVO
n = 53
n = 54
n = 73
n = 34
HR, 0.46
P = 0.005
HR, 0.37
P = 0.001
OS (%)
Months

Conclusions

- This study demonstrates the utility of combining AI-powered CD8 topology classifications with TC PD-L1 expression as a composite biomarker associated with immunotherapy response in patients with advanced melanoma
- In patients with melanoma whose tumors had PD-L1 expression < 1% treated with NIVO + IPI, median OS was significantly longer in patients with CD8-excluded tumors than in those with an inflamed phenotype
- In patients treated with NIVO + IPI, there was a trend toward a lower frequency of AEs in patients whose tumors had a PD-L1 expression < 1% and in patients with a CD8-excluded compared with a CD8-inflamed phenotype
- Use of a composite biomarker, consisting of AI-driven intratumoral CD8-topology classifications and PD-L1 expression, identified a larger subset of patients who benefited from treatment with NIVO monotherapy or NIVO + IPI, compared with classification using PD-L1 status alone
- Further studies are underway to identify mechanisms underlying responses to NIVO + IPI in the subgroups of patients analyzed in this study

References

1. Geng Y, et al. *Cell Physiol Biochem* 2015;37:1560-1571.
2. Manabe K, et al. *J Dermatol* 2021;48:1186-1192.
3. Hendry S, et al. *Adv Anat Pathol* 2017;24:235-251.
4. Desbois M, et al. *Nat Commun* 2020;11:5583.
5. Amgad M, et al. *NPJ Breast Cancer* 2020;6:16.
6. Kather JN, et al. *Elife* 2018;7:e36967.
7. Braun DA, et al. *Nat Med* 2020;26:909-918.
8. Brunnström H, et al. *Mod Pathol* 2017;1411-1421.
9. Tsao MS, et al. *J Thorac Oncol* 2018;13:1302-1311.
10. Larkin J, et al. *N Engl J Med* 2015;373:23-34.

Acknowledgments

- The patients and families who made this study possible
- The clinical study teams who participated and CheckMate 067 investigators, including F Stephen Hodi, MD, James Larkin, MD, and Jedd D. Wolchok, MD
- Dako, an Agilent Technologies, Inc. company, for collaborative development of the PD-L1 IHC 28-8 pharmDx assay
- Bristol Myers Squibb (Princeton, NJ) and Ono Pharmaceutical Company Ltd. (Osaka, Japan)
- This study was supported by Bristol Myers Squibb
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Emily Motola, PhD, and Matthew Weddig, BA, of Spark Medica Inc, funded by Bristol Myers Squibb

Presented at the Society for Immunotherapy of Cancer's 36th Annual Meeting, November 10-14, 2021.

Email: George.Lee@bms.com

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from the author of this poster.